Expressing AAT from circular RNA-encoding vectors as a promising gene therapy approach for Alpha 1-antitrypsin deficiency

Abstract # 598
Presented at ASGCT 2024

O'Leary ET[1,2], Vikberg S[1,2], Zhang J[1,2], Hjelmqvist D[1,2], Wiklund ED[1], Levitsky V[1], Hansen TB[1]

1. Circio AB, Huddinge, Sweden
2. Karolinska Institutet, CLINTEC, Huddinge, Sweden

Introduction

Alpha-1 antitrypsin deficiency (AATD) is a genetic disorder affecting the lung and liver and caused by mutations in the SERPINA1 gene, notably the Z and S variants. The Z variant, in particular, leads to toxic accumulation of mutant AAT protein, resulting in lung emphysema and liver cirrhosis. Current treatments are primarily symptomatic, with liver transplantation representing the sole curative option. However, there are promising gene therapies in development aimed at elevating AAT levels in the blood and reducing toxic accumulation in the liver, potentially slowing disease progression.
Circio is pioneering an alternative therapeutic approach leveraging our proprietary vectorized circular RNA (circRNA) expression platform, **circVec**. CircRNA boasts enhanced intra-cellular stability compared to mRNA, resisting exo-nucleolytic decay. It's emerging as a promising therapeutic modality for disorders necessitating high and sustained transgene expression, such as AATD.

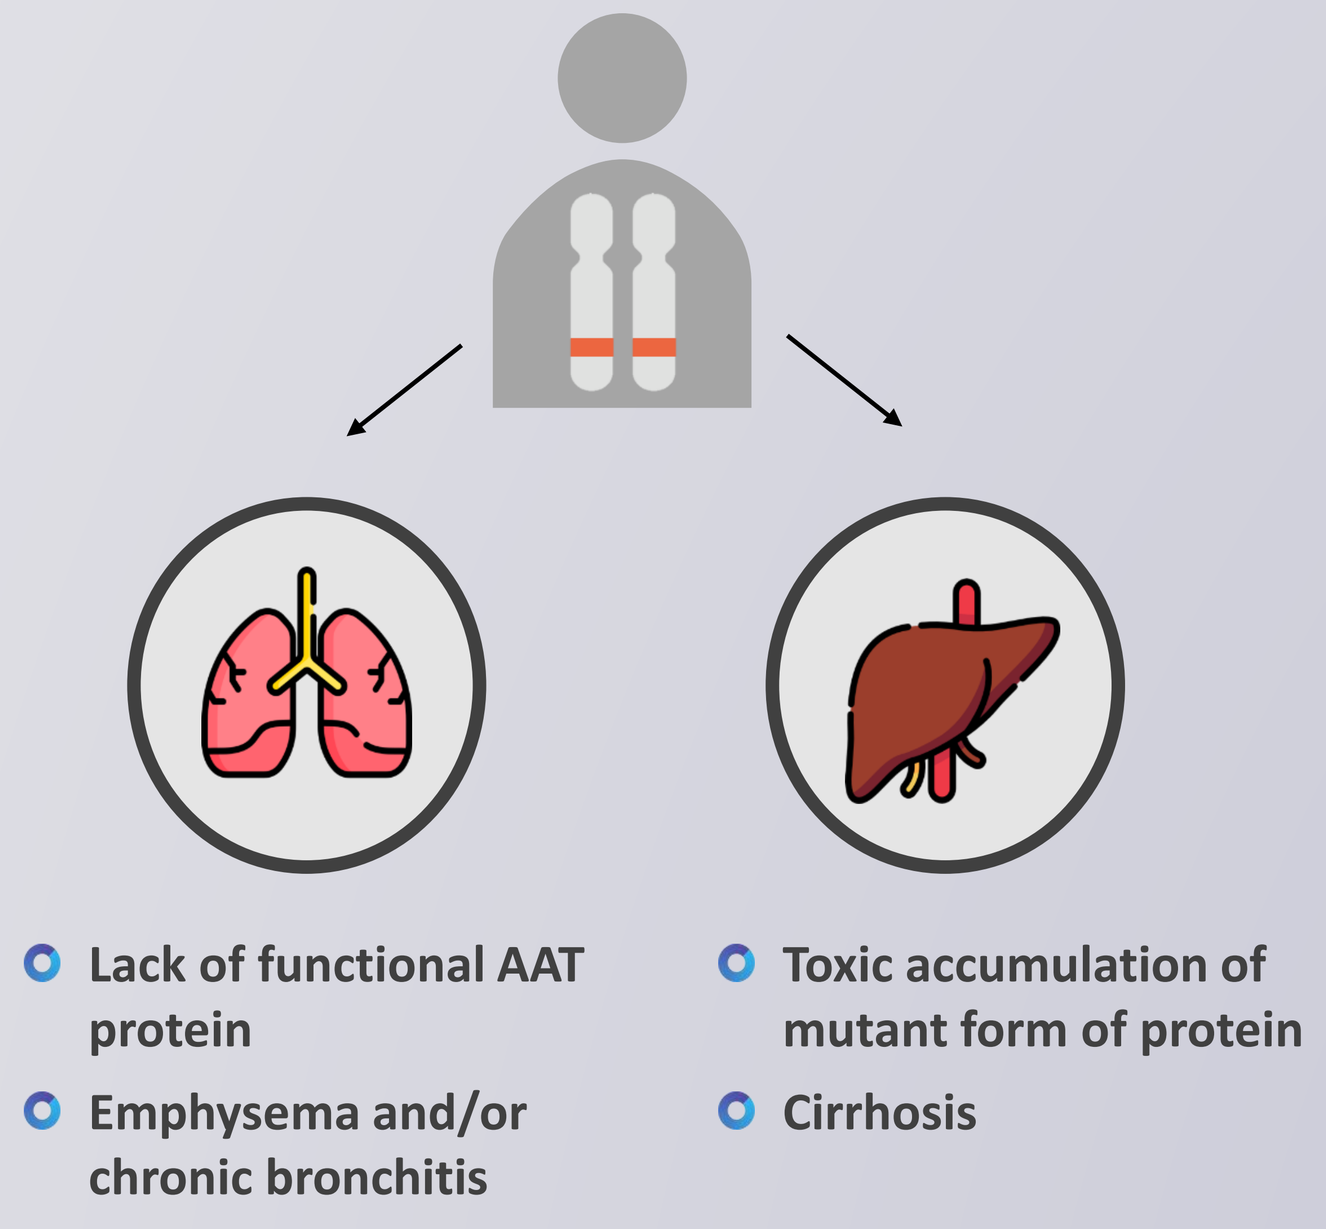

Lack of functional AAT protein
Emphysema and/or chronic bronchitis
Toxic accumulation of mutant form of protein
Cirrhosis

circVec cassette design is crucial for high-yield AAT expression

CircVec, an innovative circRNA expression platform, utilizes intracellular splicing machinery for in situ circRNA generation (**Fig. 1A**). By optimizing circular RNA biogenesis and cap-independent translation, circVec development has enhanced protein yield significantly, as detailed in **Poster #1713**. Here, we employ the circVec system for AAT expression, assessing various cassette iterations. CircVec2.1 achieves the highest AAT production levels (**Fig. 1B**), demonstrating comparable expression to conventional mRNA at early timepoints, despite lower circRNA copy numbers (**Fig. 1C**). Concurrently, we explore codon optimization (CO) to improve circVec-mediated translation. Our in-house "Cio1" algorithm generates RNA variants which yield higher AAT levels compared to the wildtype or other RNAs produced based on existing CO algorithms (**Fig. 1D**).

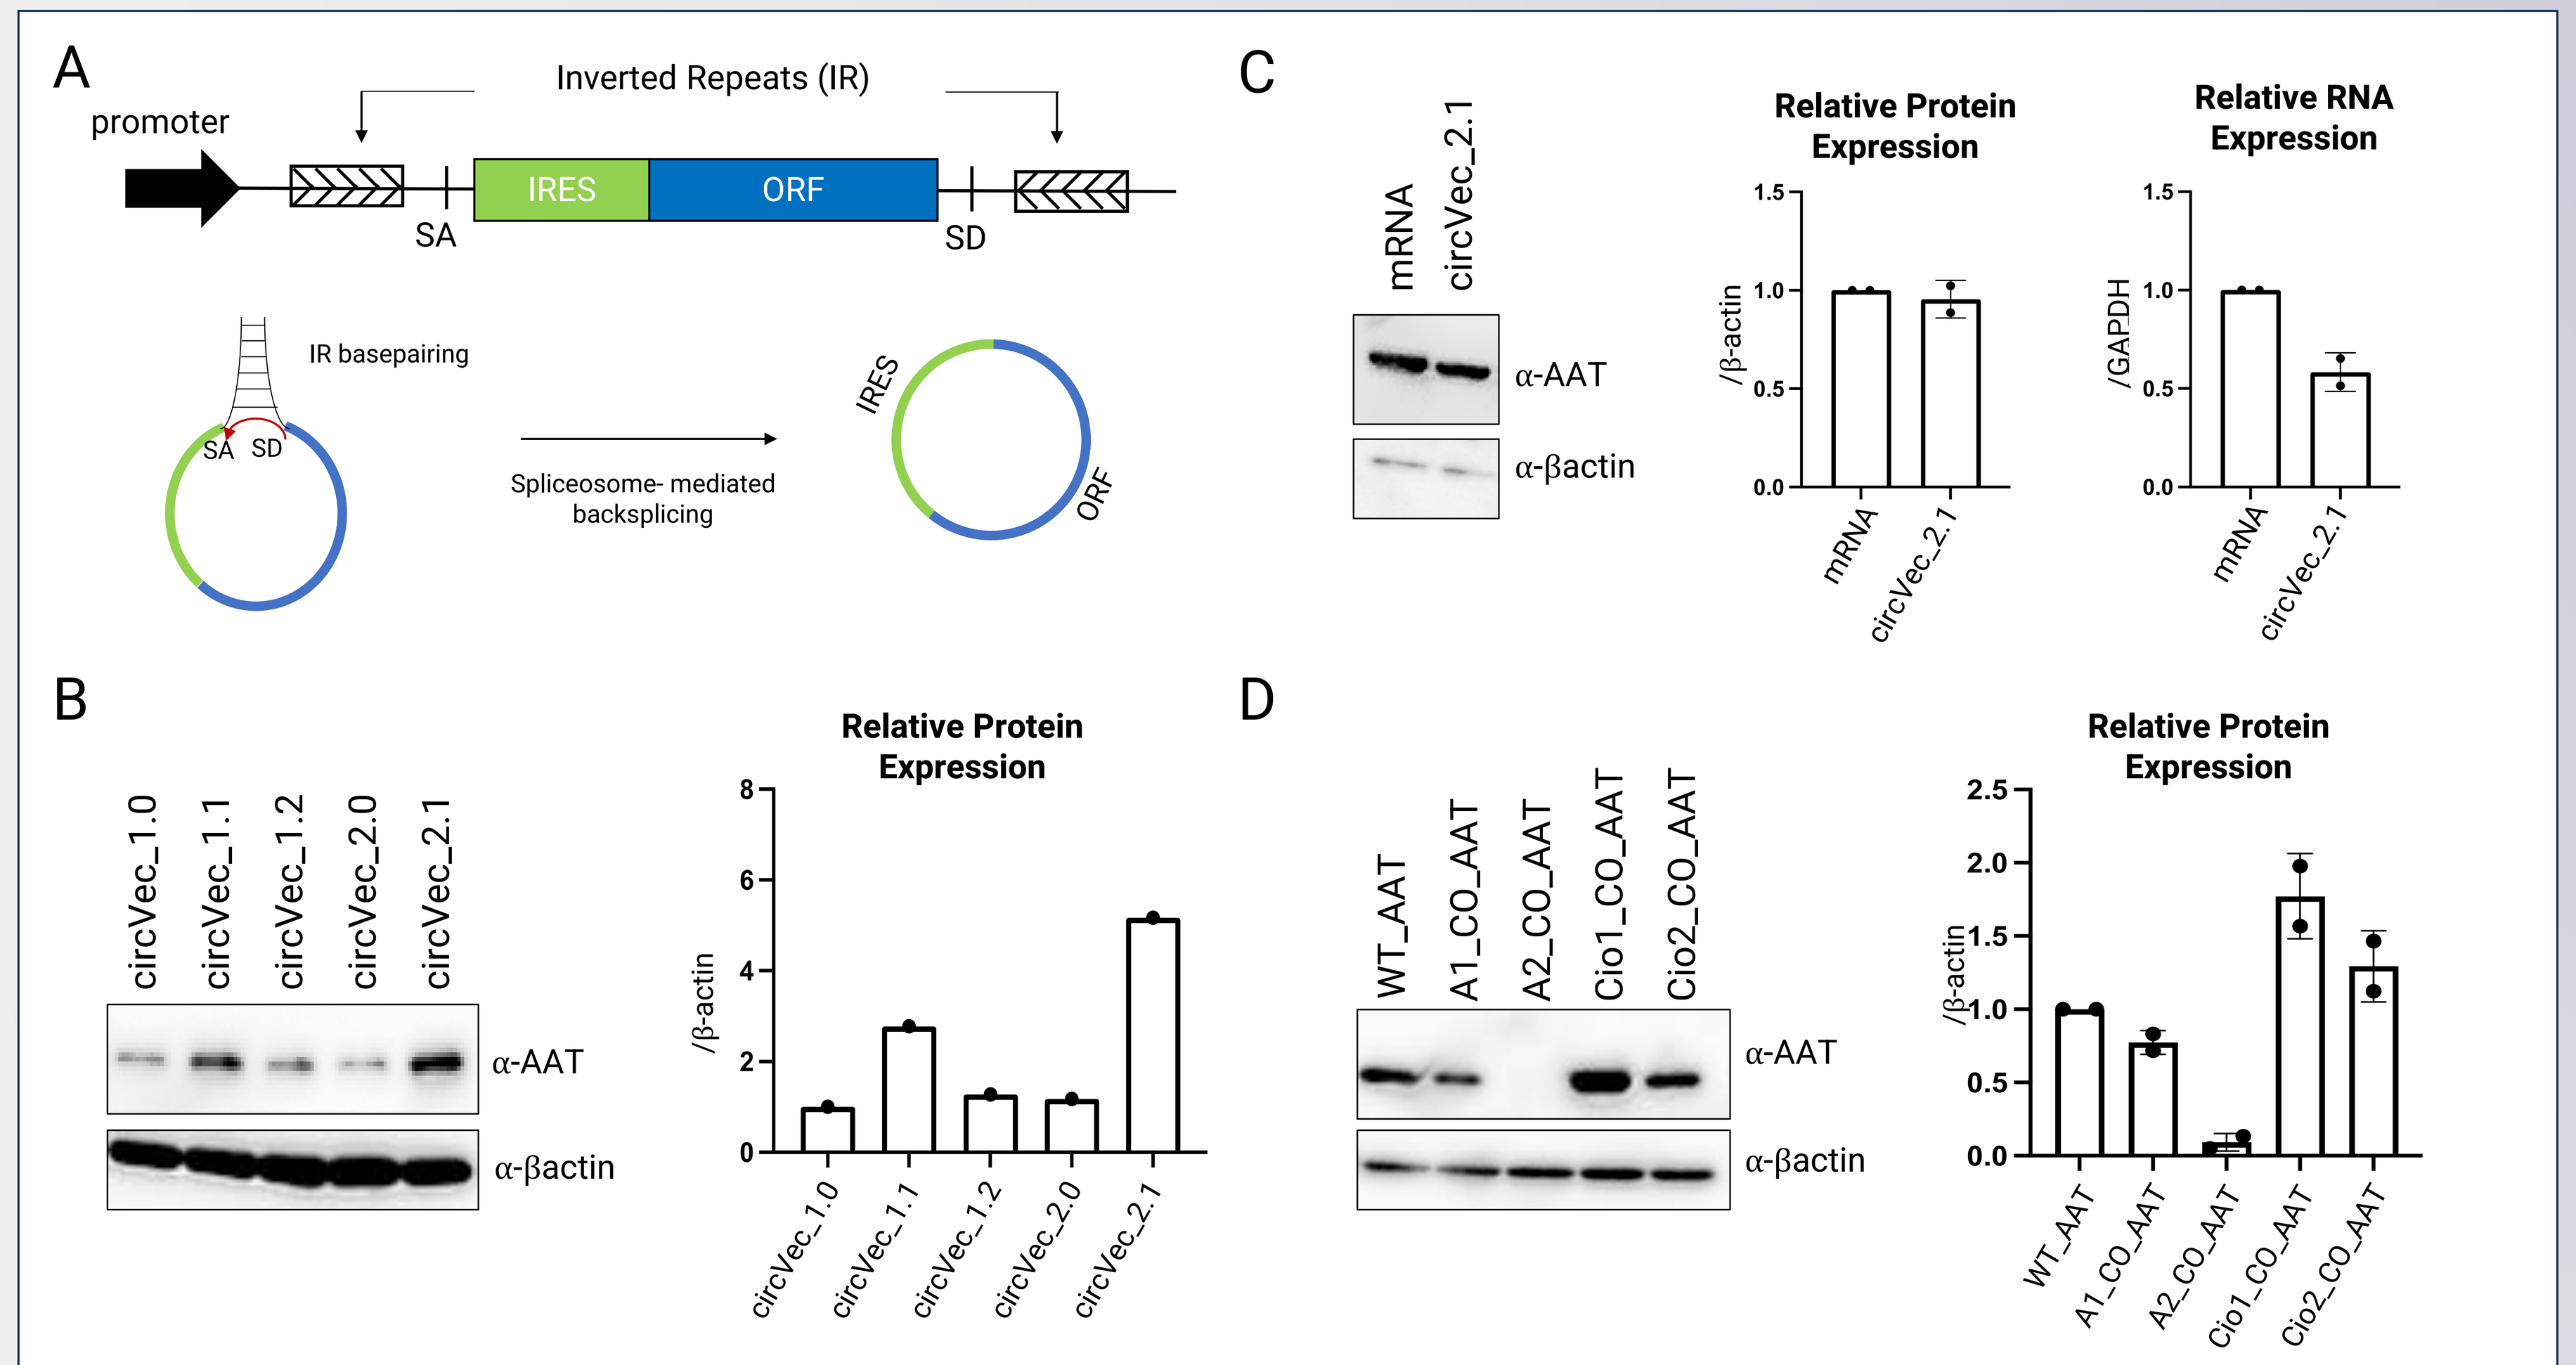

A
Inverted Repeats (IR)
promoter
IRES
ORF
SA
SD
IR basepairing
Spliceosome- mediated backsplicing
B
circVec_1.0
circVec_1.1
circVec_1.2
circVec_2.0
circVec_2.1
α-AAT
α-βactin
Relative Protein Expression
/β-actin
C
mRNA
circVec_2.1
Relative RNA Expression
/GAPDH
D
WT_AAT
A1_CO_AAT
A2_CO_AAT
Cio1_CO_AAT
Cio2_CO_AAT

Figure 1: circVec cassette design is crucial for high-yield circRNA expression. A) Schematic of circVec cassette design and spliceosome-mediated circRNA biogenesis. **B)** Western blot analysis of AAT protein expression from HepG2 cells transfected with different iterations of the circVec cassette. CircVec 1.1 and 1.2 have optimized inverted repeat (IR) elements and circVec 1.x vs 2.x utilize different IRES elements. **C)** AAT protein ad RNA levels from HEK293T cells transfected with a conventional mRNA expression vector or circVec, were assessed by western blotting and RT-qPCR respectively, n=2. **D)** Western blot analysis of AAT protein expression from HepG2 cells transfected with different codon optimised (CO) versions of the AAT ORF. A1 & A2 correspond to codon optimization achieved through two established algorithms, while "Cio" signifies AAT codon optimization achieved through Circio's proprietary algorithm

Enhanced circRNA stability confers prolonged AAT protein expression

Almost all cellular RNA turnover is facilitated by exonucleolytic decay. CircRNAs are devoid of 5' and 3' ends and thereby resistant to exonucleases. Consequently, circRNA exhibits high stability compared to mRNA. Here, we compared AAT expression from circVec_1.0 with mRNA expression from the same backbone (**Fig. 2A**) in HepG2 and C2C12 cell lines. The inherent stability of circRNA results in AAT protein accumulation over time, whereas mRNA levels decline more rapidly (**Fig. 2B-C**), suggesting that the circRNA-based vectors extend the duration of protein expression.

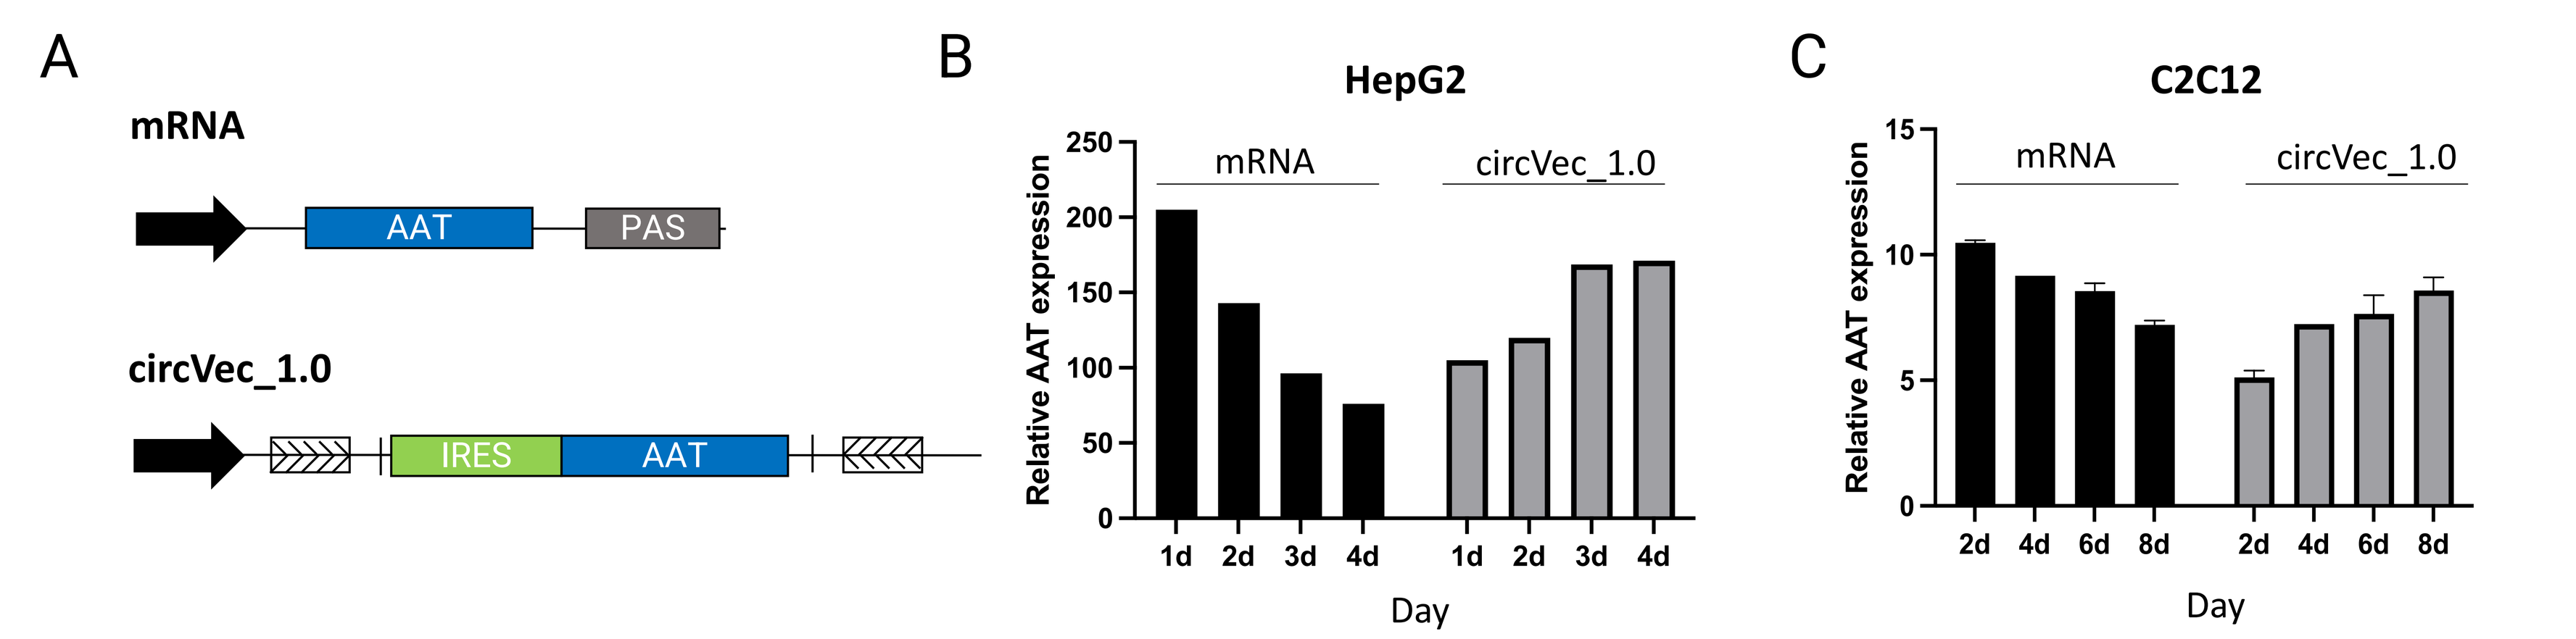

A
mRNA
AAT
PAS
circVec_1.0
IRES
AAT
B
HepG2
C
C2C12
mRNA
circVec_1.0
Relative AAT expression
Day

Figure 2: Superior circRNA stability enhances durability protein expression. A) Schematic of conventional mRNA expression cassette (upper) and circVec_1.0 circRNA expression cassette (lower). **B-C)** AAT protein expression was assessed by ELISA in HepG2 (B) and C2C12 (C) cells, over 4 and 8 days, respectively. Protein levels were normalised to empty vector control.

Efficient depletion of pathogenic AAT variants by Circio remove vector

AATD stems from SERPINA1 gene mutations, notably the Z (Glu342Lys) and S (Glu264Val) variants, with 95% of severe cases showing homozygosity for the Z allele mutation. We introduce a vector-based system encoding a remove molecule for sequence-specific depletion of these variants (**Fig. 3A**). To reduce off-target depletion of circVec-encoded AAT, we codon-optimized the target regions within circAAT (**Fig. 3B**). Two remove vector (REM) designs, REM_1 and REM_2, target each variant, with REM_1 showing superior knock-down efficiencies for both variants (>90%) compared to the empty vector control (>90%) and REM_2 (**Fig. 3C**). Although wild-type AAT is partially reduced, mutant AAT is significantly more impacted by REM expression, while circAAT exhibits high REM resistance (**Fig. 3D**).

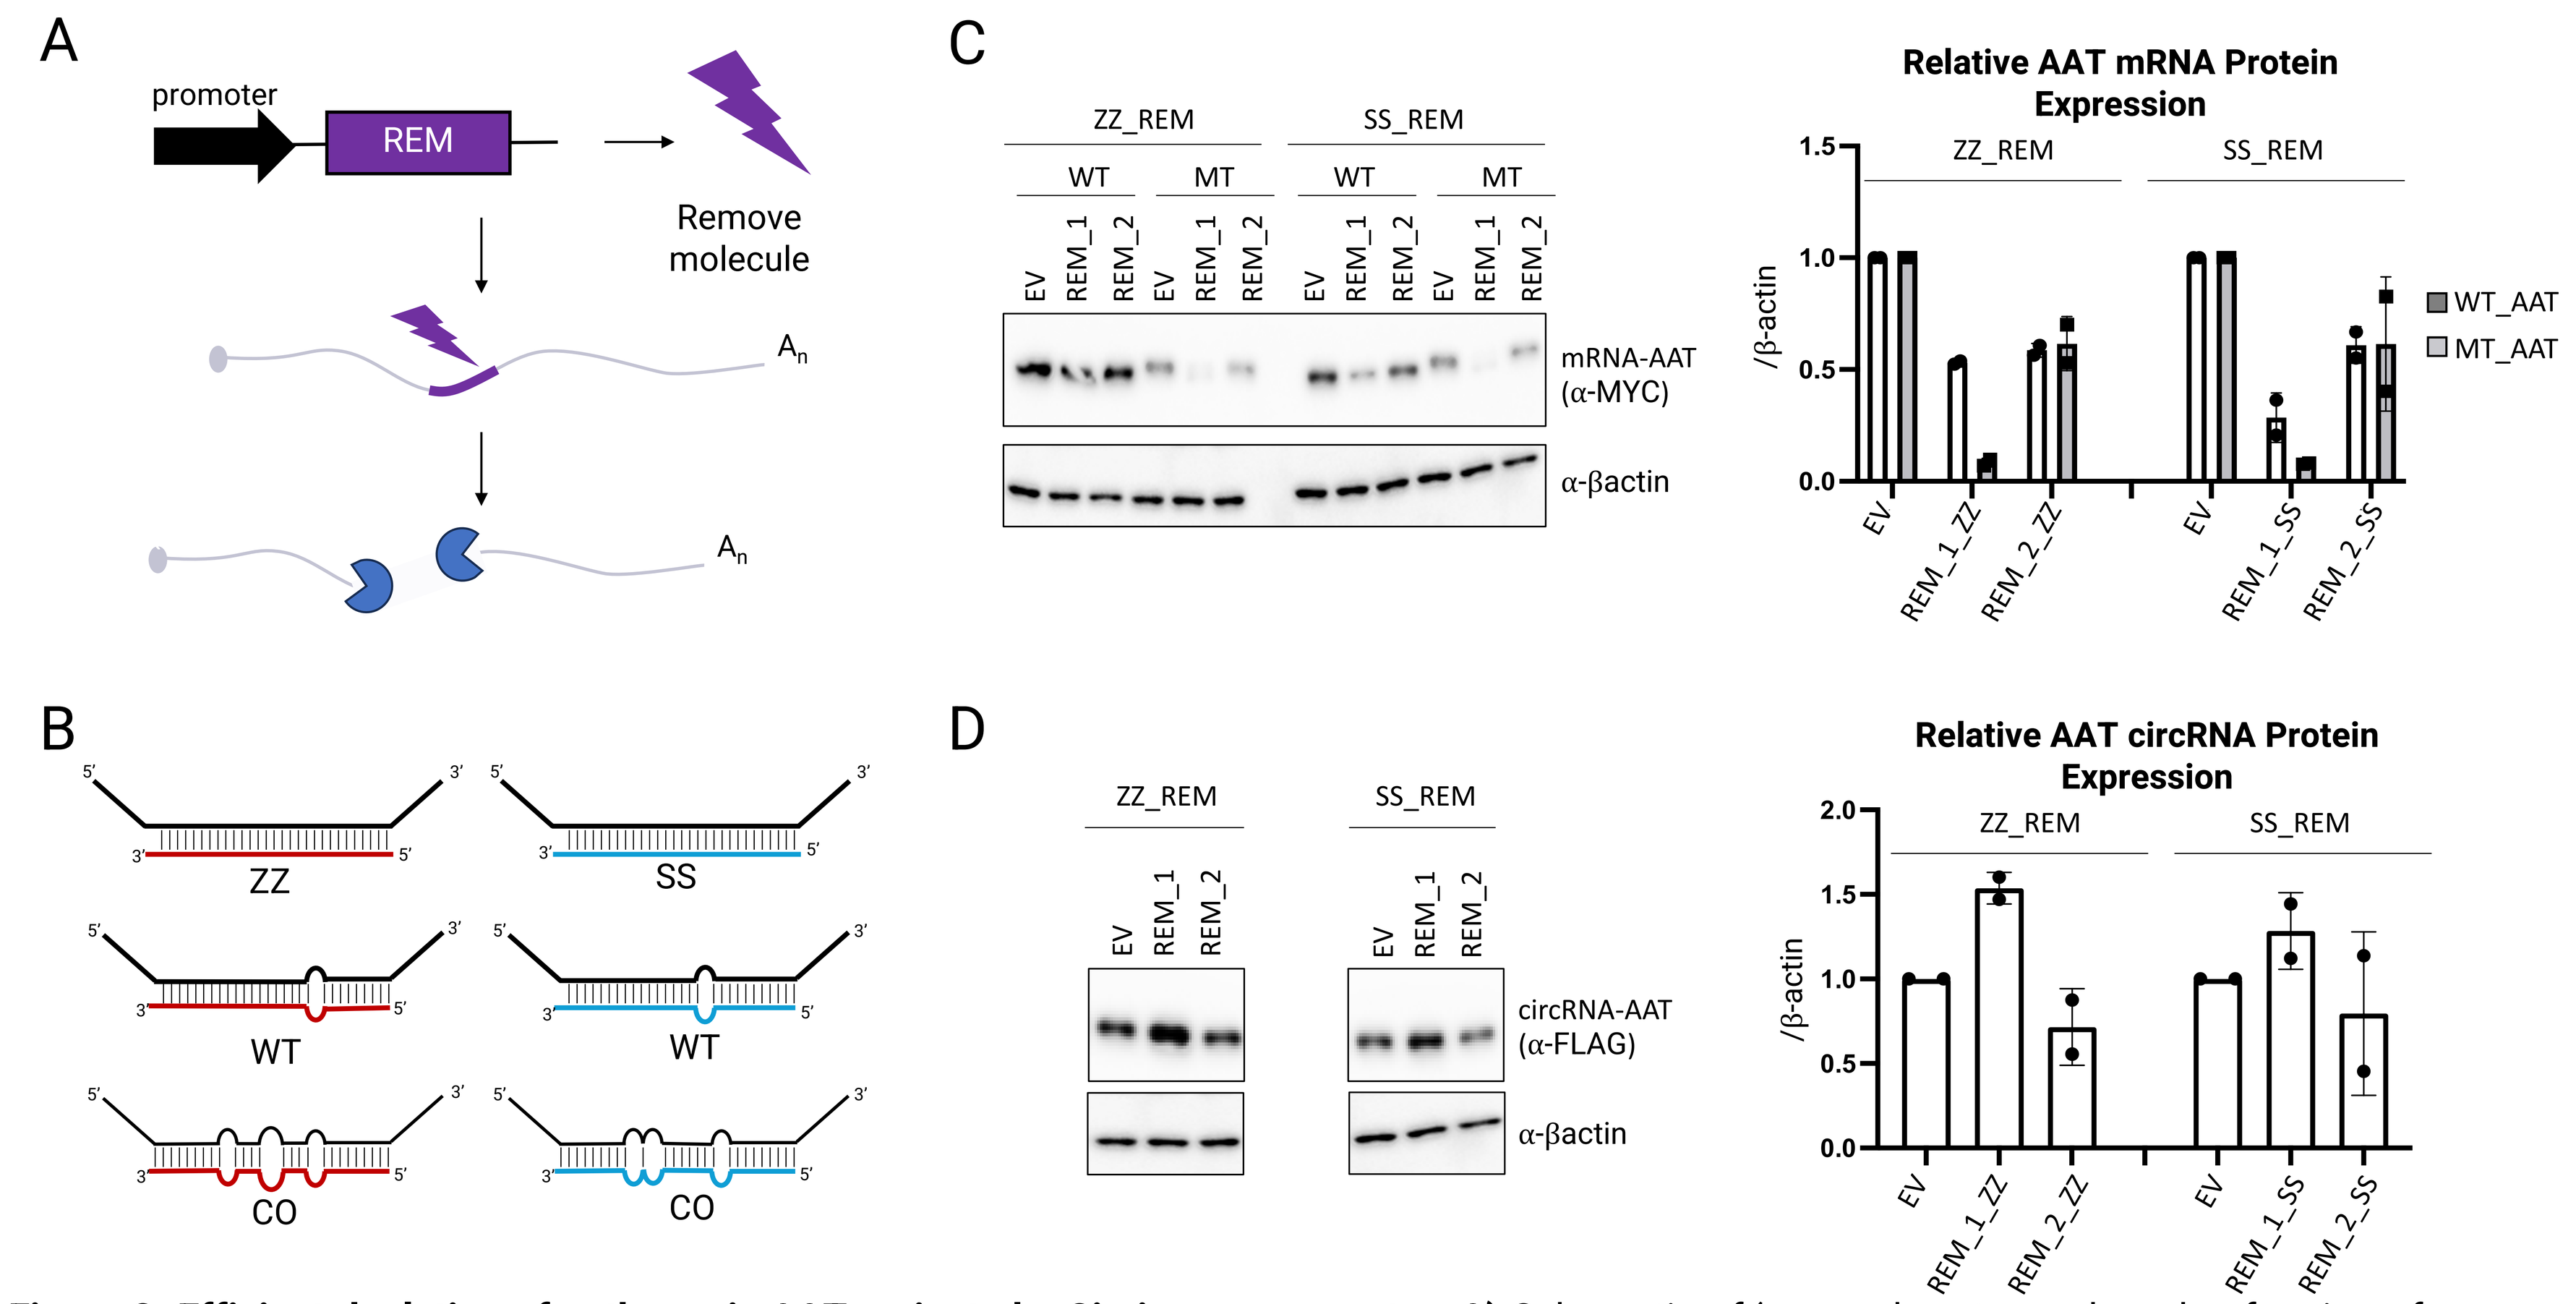

A
promoter
REM
Remove molecule
B
ZZ
SS
WT
CO
C
ZZ_REM
SS_REM
EV
REM_1
REM_2
mRNA-AAT (α-MYC)
α-βactin
Relative AAT mRNA Protein Expression
WT_AAT
MT_AAT
/β-actin
D
circRNA-AAT (α-FLAG)
Relative AAT circRNA Protein Expression

Figure 3: Efficient depletion of pathogenic AAT variants by Circio remove vector. A) Schematic of 'remove' vector and mode of action of remove molecule. **B)** Schematic of remove molecule targeting ZZ (red) and SS (blue) variant. Bulges in sequence represent mismatch regions between remove molecule and target RNA; single mismatch between remove molecule and wildtype (WT) RNA. Several mismatches introduced between remove molecule and codon optimised (CO) RNA sequence. **C)** Knockdown efficiency of remove vector was assessed by western blot analysis of AAT protein expression from HEK293T cells co-transfected with WT or mutant (MT) AAT reporter and the indicated REM or EV control vector, at a ratio of 1:4. **D)** Resistance of circAAT to the remove molecule was assessed by western blot analysis of HEK293T cells co-transfected with circVec and the indicated REM or EV control vector, at a ratio of 1:4. β-actin was used as loading control; n=2 biological replicates.

Bifunctional circVec successfully depletes pathogenic AAT variants and replenishes functional AAT

Integrating the REM depletion system into the circVec platform, a bimodal vector with the capability to target both lung and liver-associated AATD pathologies has been developed, coined 'Remove'n'Replace' (**Fig. 4A-B**). This technology effectively depletes pathogenic AAT variants while simultaneously expressing functional AAT. Here, we present a proof-of-concept experiment demonstrating the efficacy of this system *in vitro* (**Fig. 4C-D**).

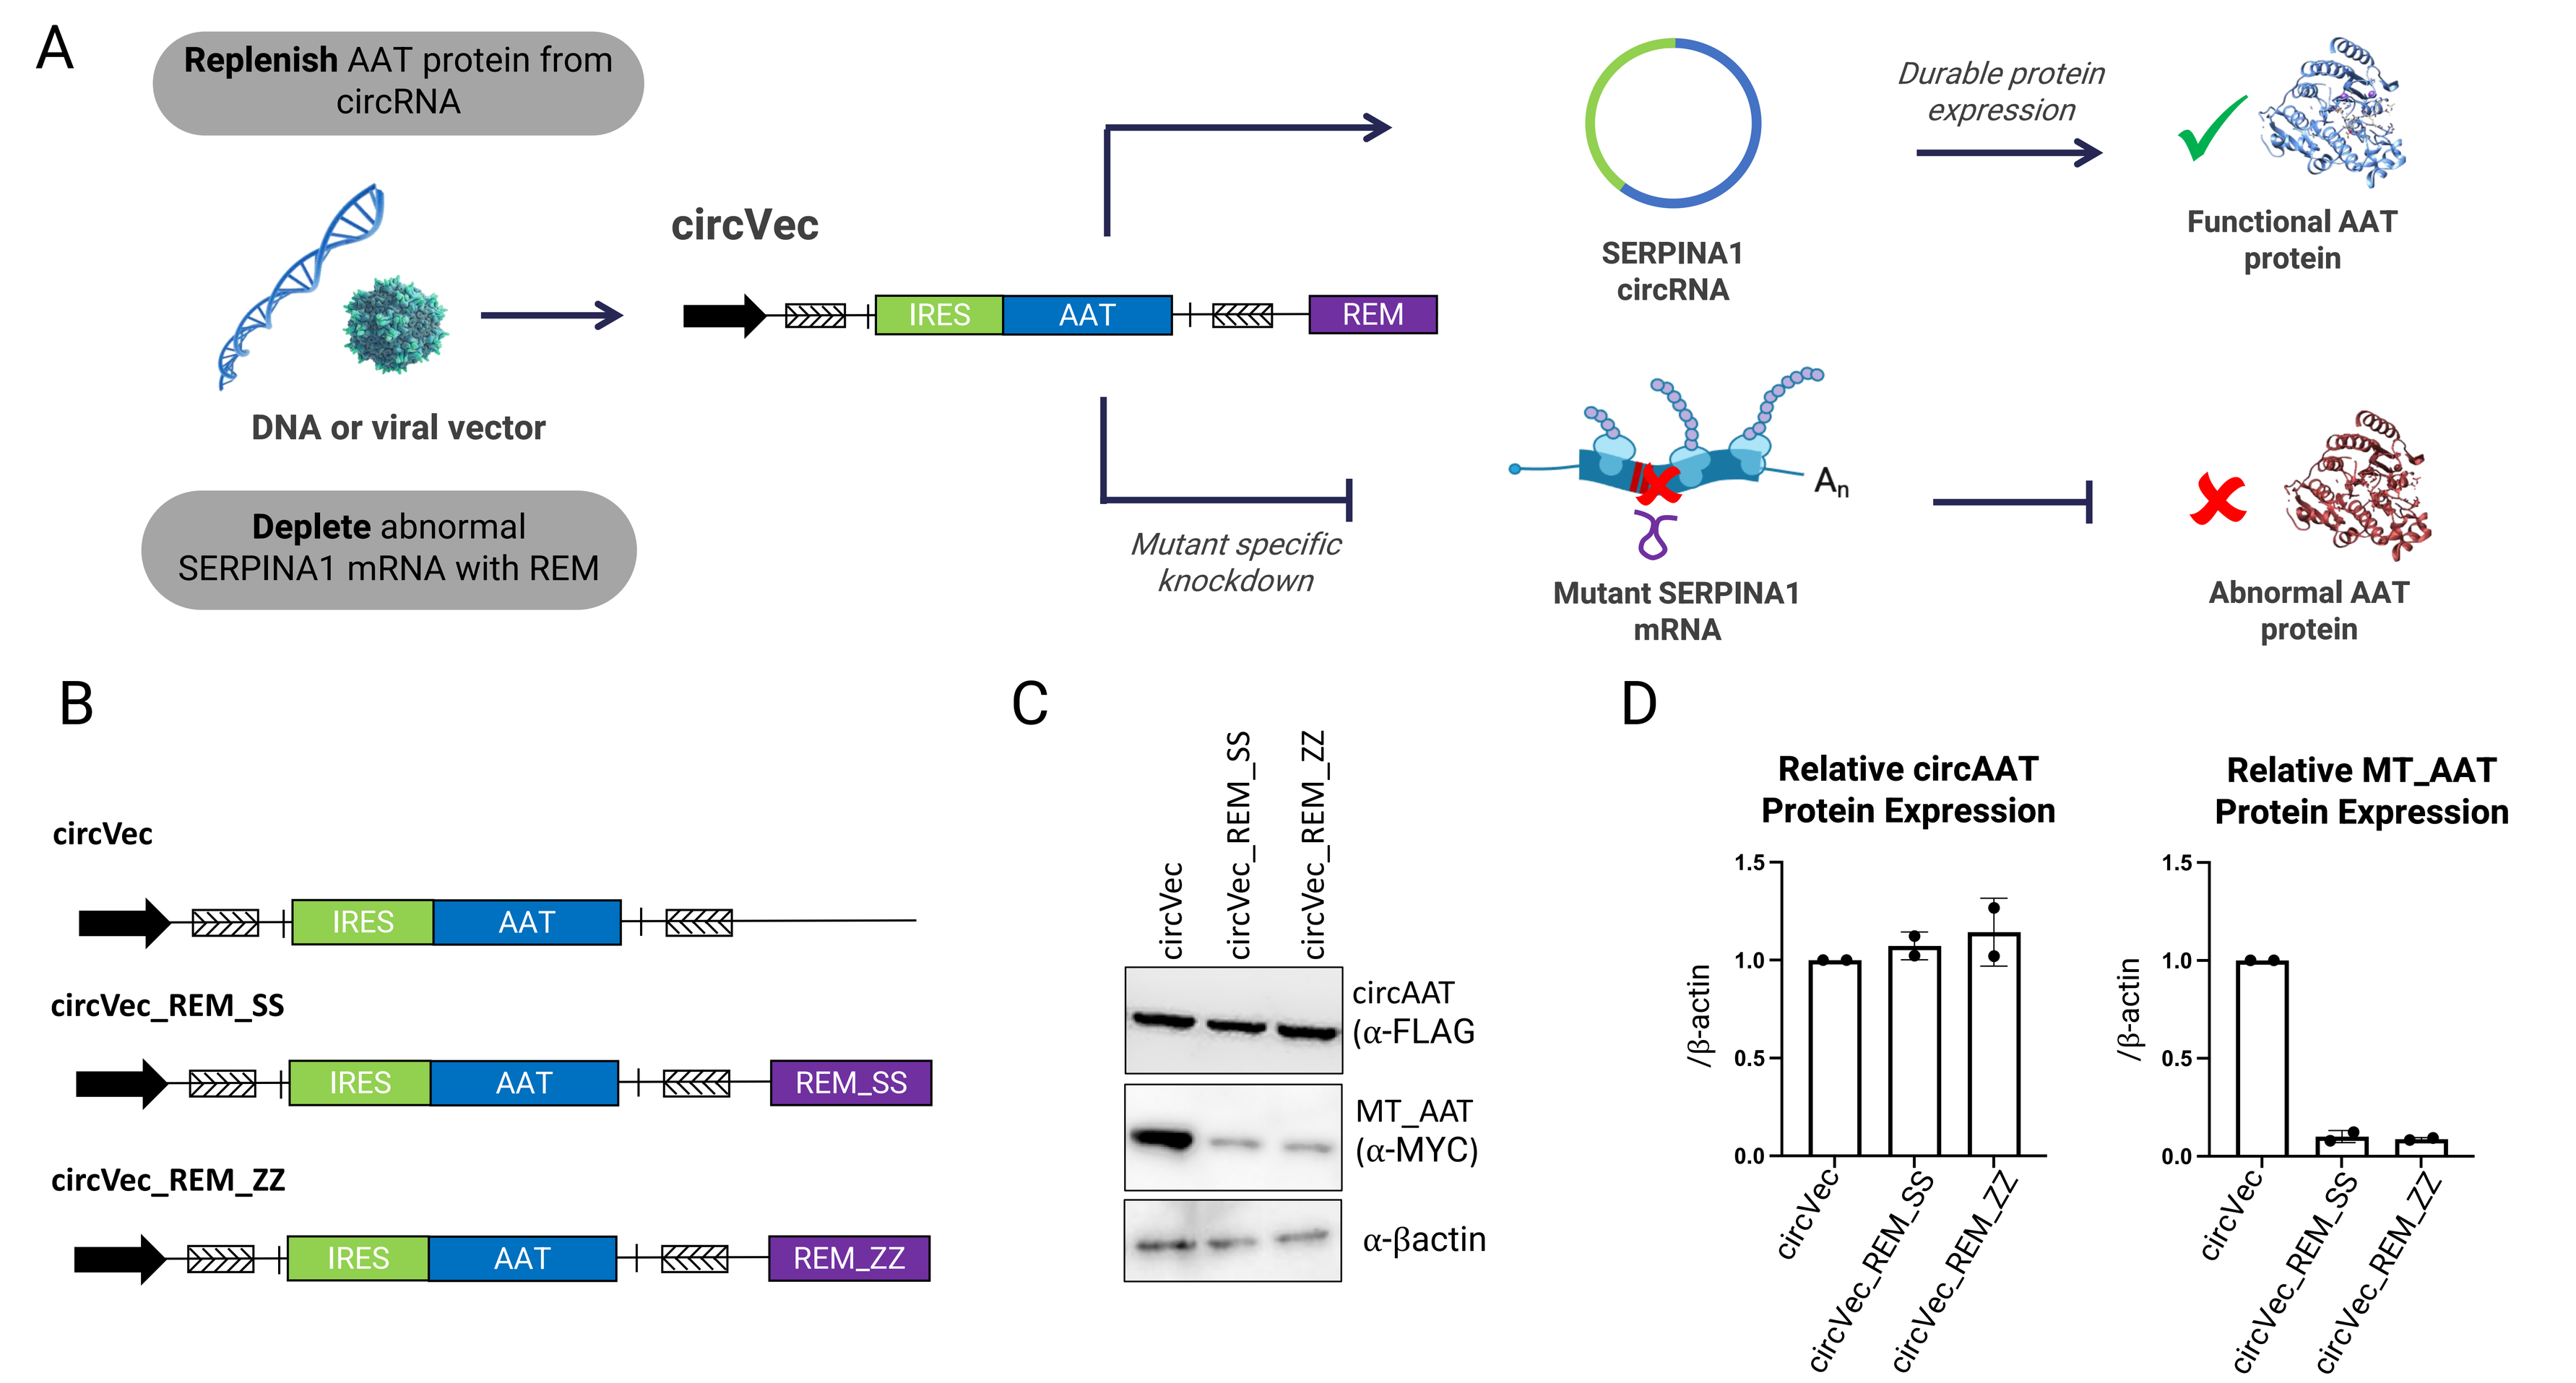

A
Replenish AAT protein from circRNA
DNA or viral vector
Deplete abnormal SERPINA1 mRNA with REM
circVec
IRES
AAT
REM
SERPINA1 circRNA
Durable protein expression
Functional AAT protein
Mutant specific knockdown
Mutant SERPINA1 mRNA
Abnormal AAT protein
B
circVec
circVec_REM_SS
circVec_REM_ZZ
REM_SS
REM_ZZ
C
circAAT (α-FLAG
MT_AAT (α-MYC)
α-βactin
D
Relative circAAT Protein Expression
Relative MT_AAT Protein Expression
/β-actin

Figure 4: circVec remove'n'replace technology successfully depletes pathogenic AAT variants while replenishing functional AAT. A) Schematic of remove'n'replace circVec concept. **B)** Schematic of circVec (upper), circVec_REM_SS (middle) and circVec_REM_ZZ (lower) cassette. **C)** HEK293T cells were co-transfected with indicated circVec and mutant AAT reporter (MT_AAT), at a ratio of 2:1. Wildtype AAT (from circRNA) and mutant AAT were assessed by western blotting using FLAG and MYC antibodies, respectively, and a loading control (β-actin). **D)** Relative wildtype AAT expression from circVec (left side) and mutant AAT (right side); n=2 biological replicates.

Conclusions

1) CircVec 'Remove'n'Replace' vector enables the combined treatment of both lung and liver associated AATD pathologies.
2) Successful remove vector for depletion of pathogenic AAT variants established.
3) Superior circRNA stability facilitates prolonged AAT protein expression.
4) CircVec cassette design is crucial for high-yield circAAT expression.

The results support further development of circRNA-based platforms in the treatment of a variety genetic diseases and, suggest that circRNA could replace mRNA as the expression platform of choice for vector-based therapies.

For more information on Circio's platform development visit poster # 1713.

Acknowledgements

This study was funded by Circio Holding ASA Norway. The authors thank everyone in the company for a great atmosphere and working environment.

Contact information

Copies of this poster can be obtained via the QR code (for personal use and may not be reproduced without author permission)

Eoghan.oleary@circio.com